AACR 2026
Session PO.CTP01.03
Poster CT220/15

A Phase 1b/2 Study on the Efficacy and Safety of BXQ-350 in Combination with mFOLFOX7 and Bevacizumab in Newly Diagnosed Metastatic Colorectal Cancer (mCRC) Patients

R. Patel [1], D. Flora [2], F. Lee[3], A. Pimentel[4], J. Gemmill[5], D. Sohal [6], V. Sharma[7], S. Sharif [8], N. Gabrail[9], K. Chung[10], J. Villano[11], D. Outlaw[12], A. Baron[13], B. Boulmay [14] G. Tapolsky [15], J. Beach[15], M. Gazda[15], T. Arshad[15]

Background

Sphingolipids are bioactive signaling molecules implicated in cancer

- **Ceramides** are pro-apoptotic and promote an anti-tumoral immune environment
- **Sphingosine-1-phosphate** (S1P) promotes tumor progression and a pro-tumoral immune environment
- **Several studies have shown elevated ceramide or low S1P levels** are associated with **improved survival and better prognosis in CRC patients**

BXQ-350 is a nanovesicle formulation of Saposin C, an allosteric activator of sphingolipid metabolism

- Normalizes dysregulated sphingolipid metabolism, lowering S1P and increasing ceramide levels

BXQ-350 was investigated in a Phase 1 study in all-comer cancer patients with recurrent Malignancies (NCT02859857)

- BXQ-350 was **safe and well-tolerated**

BXQ-350 + mFOLFOX7 & Bevacizumab study design:

Phase 1b/2 study in combination with mFOLFOX7 and Bevacizumab in newly diagnosed mCRC patients

- **Phase 1b/2:**
 - Safety dose escalation to establish RP2D. Patients to start at 1.8 mg/kg BXQ-350 in combination with mFOLFOX7 and Bevacizumab; if no MTD, BXQ-350 increased to 2.4 mg/kg which would be the RP2D (if no MTD).
 - 30 patient expansion cohort at the RP2D
- **Primary objectives Phase 1b/2:**
 - Select **RP2D** (safety profile, DLTs)
 - **Preliminary efficacy** of the combination based on **ORR (Overall Response Rate), (Cumulative Oxaliplatin Dose)**
- **Secondary objectives Phase 1b/2:**
 - Overall safety and tolerability of combination
 - Efficacy of BXQ-350 + mFOLFOX7 & Bevacizumab based on **DCR (Disease Control Rate), OS (Overall Survival), PFS (Progression Free Survival)**
 - Assess whether BXQ-350 decreases development, intensity or duration of CIPN based on neuropathy scores from EORTC questionnaires (QLQ-C30 and CIPN20)

University of California, Irvine, CA [3] University of Miami, Miami, FL [4] Stony Brook University Hospital, New York, NY [5] University of Cincinnati, Cincinnati, OH [6] University of Louisville, Louisville, KY [7] University of Iowa, Iowa City, IA [8] Gabrail Cancer Center, Canton, OH [9] PRISMA Health Cancer Institute, Boiling Springs, SC [10] University of Kentucky Markey Cancer Center, Lexington, KY [11] University of Alabama at Birmingham, Birmingham, AL [12] Pacific Hematology Oncology, San Francisco, CA [13] Louisiana State University Health New Orleans, New Orleans, LA [14] Bexion Pharmaceuticals, Covington, KY [15]

Summary

- BXQ-350 is a novel biologic and a nanovesicle formulation of Saposin C, an allosteric activator of enzymes involved in sphingolipid metabolism.
- BXQ-350 is well-tolerated and showed signs of single agent activity across 20 refractory solid tumors types in a Phase 1 study of 87 patients.
- Biomarkers based on plasma levels of S1P & Ceramide, Cytokines, PBMCs, and NfLs.

Current Status

- Enrollment complete and in active follow-up
- Total of 32 evaluable patients enrolled
- 30 patients (94%, all but 2 PD patients) achieved at least 8 cycles of oxaliplatin; 33% achieved at least 12 cycles.
- **94% of patients received > 6 cycles of oxaliplatin which is significantly higher than historical controls,** 68% of patients ≥ 6 cycles of oxaliplatin (TRIBE and TRIBE 2 pooled analysis, R. Moretto *et al.*, Eur J Cancer 2025, 115470)
- **Disease control rate (DCR) is 91%, ORR is 61%* and mPFS is currently 10.6m**
- Safety profile for the combination appears better than historical safety data for mFOLFOX7 + Bevacizumab, including **reduced incidence and/or severity of CIPN.**

Related Posters at AACR 2026:

- **Session PO.CTP01.01 Poster CT084/15** A Phase 1b/2a study to evaluate the efficacy and safety of BXQ-350 in combination with mFOLFOX7 and bevacizumab in newly diagnosed metastatic colorectal carcinoma patients (mCRC): Evidence of lower incidence and severity of CIPN events.
- **Session CL01.04 Poster 3737/9** BXQ-350: A novel biologic that targets dysregulated sphingolipid metabolism and normalizes key anti-tumoral and pro-tumoral sphingolipids in newly diagnosed metastatic colorectal carcinoma patients (mCRC).
- **Session PO.PR01.04 Poster 3616/2** BXQ-350 reduces incidence, severity, and time to on-set of chemotherapy induced peripheral neuropathy via modulation of sphingolipid metabolism.

Acknowledgement: Patients who participated in the trials and their families, clinicians and staff at investigational sites, and Bexion personnel.

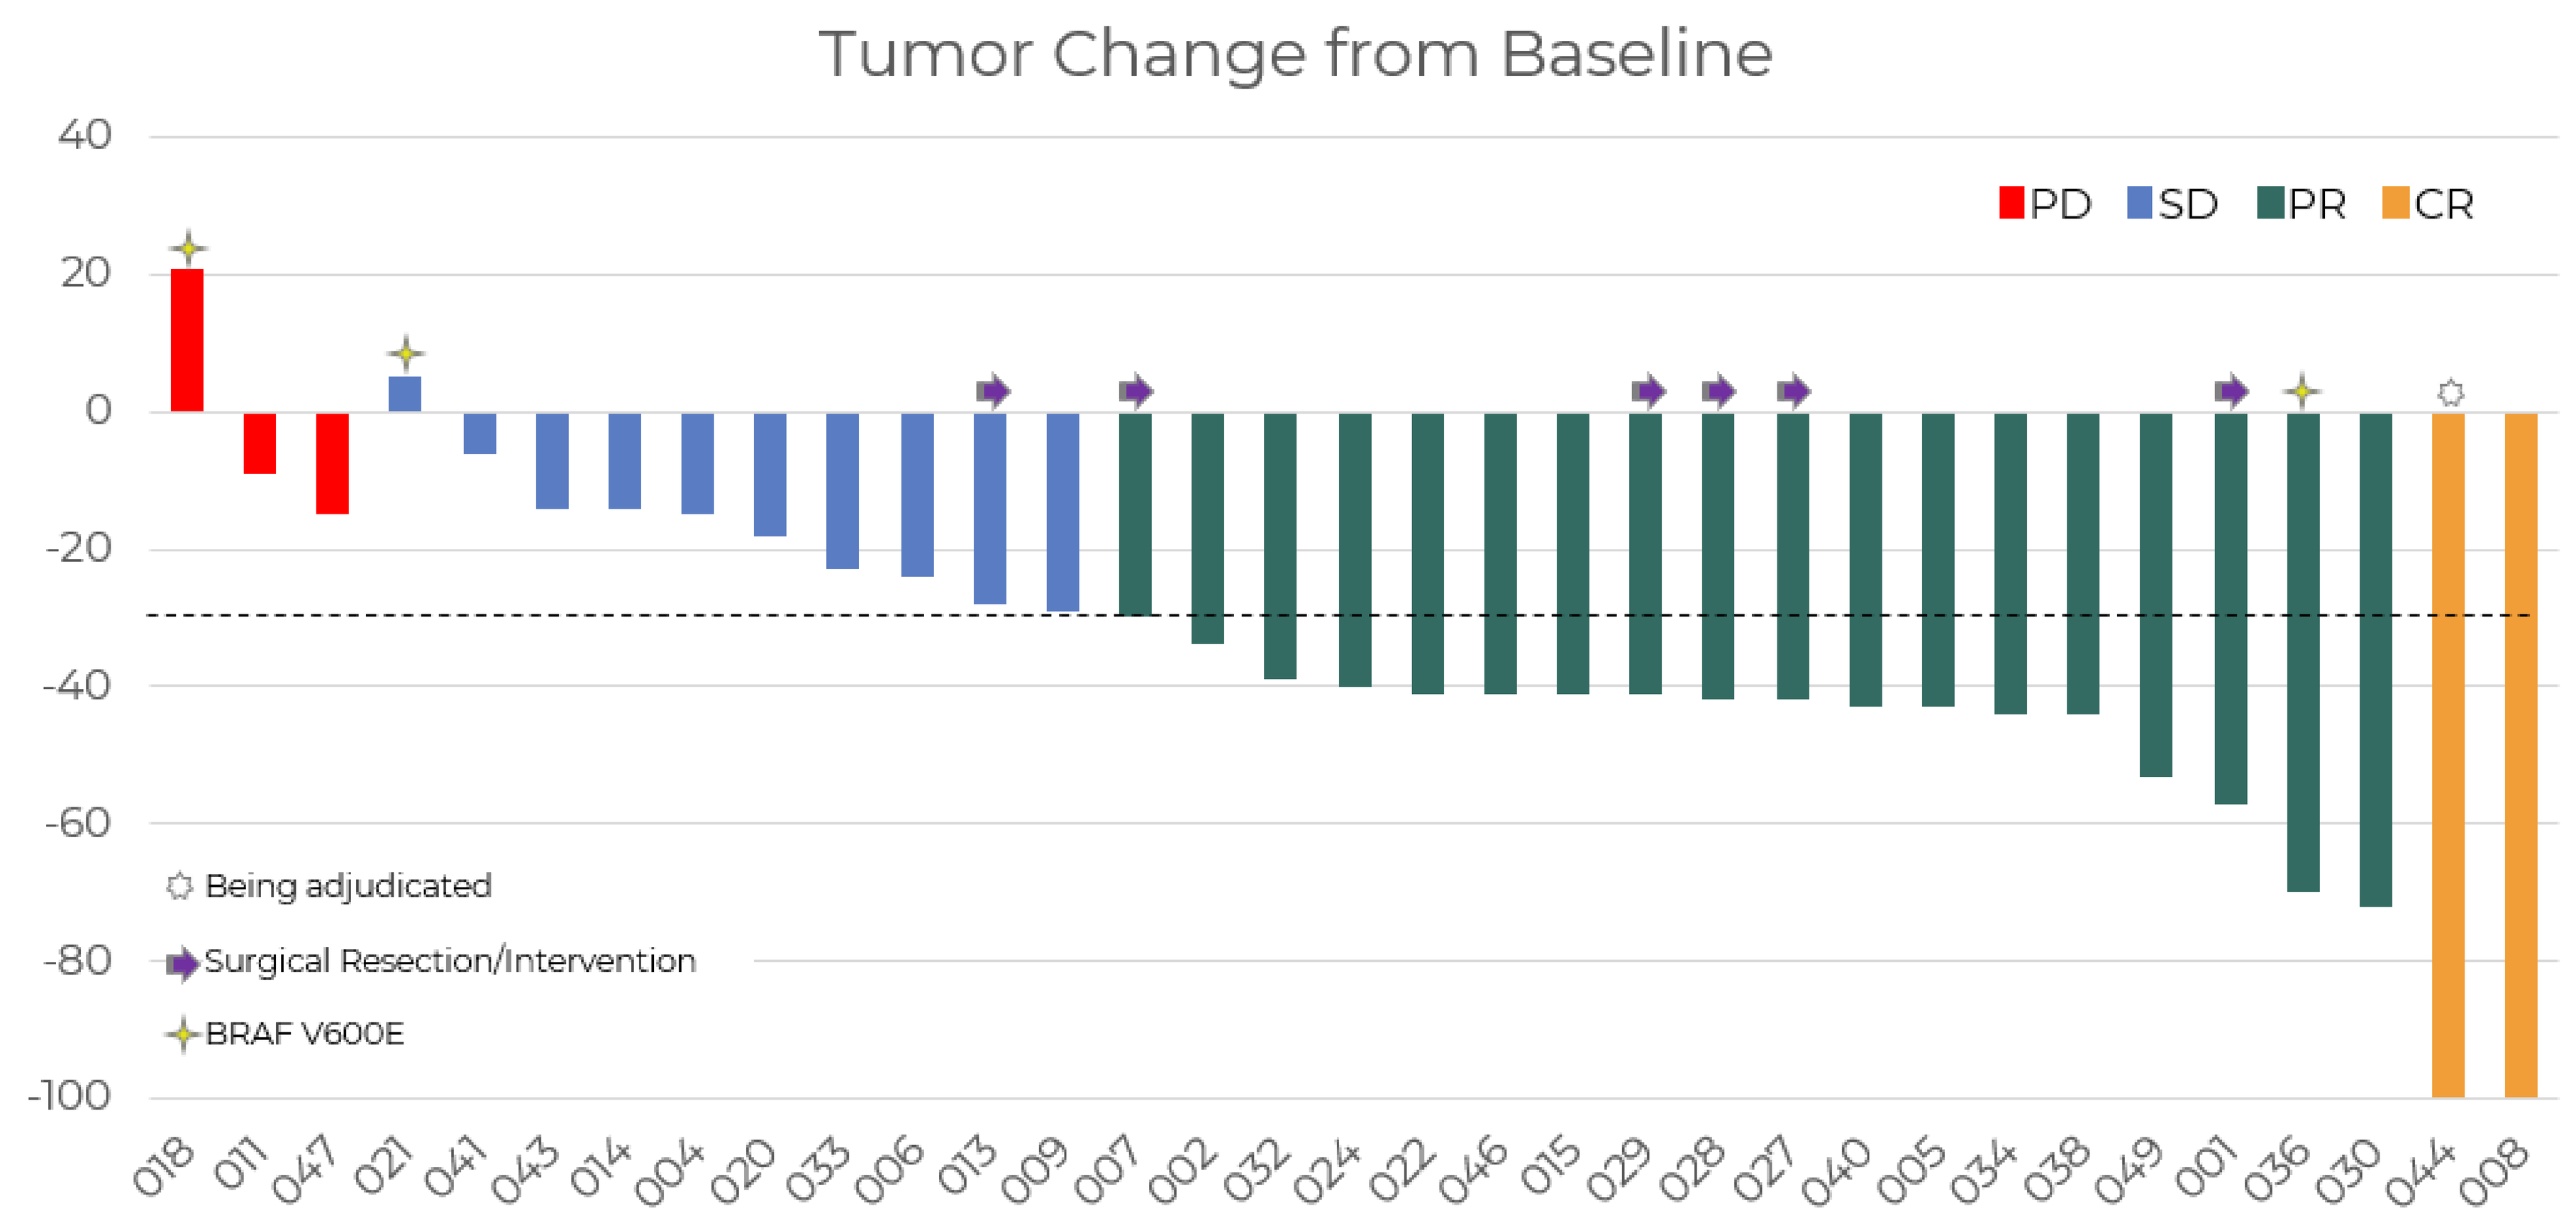

Tumor Change from Baseline
PD SD PR CR
40
20
0
-20
-40
-60
-80
-100
Being adjudicated
Surgical Resection/Intervention
BRAF V600E
018 011 047 021 041 043 014 004 020 033 006 013 009 007 002 032 024 022 046 015 029 028 027 040 005 034 038 049 001 036 030 044 008

ORR 61% *
DCR 91%
mPFS 10.6 months

- 5 of these patients left study to receive surgical resection of their tumors
- 2 additional patients left study early for surgical resection (1 PR, 1 SD, both not evaluable)
- 1 patient (029) received radioembolization treatment of liver lesion

The data are provisional and are subject to change based on future updates.

TRIBE2 Study	6+ Doses	<6 Doses
% Achieving	68%	32%
Progression Free Survival	10.1 months	8.8 months

Cremolini et al, European Journal of Cancer (2025) – 334 patients

Bexion Study	6+ Doses	<6 Doses
% Achieving	94%	6%
Progression Free Survival	10.6 months	

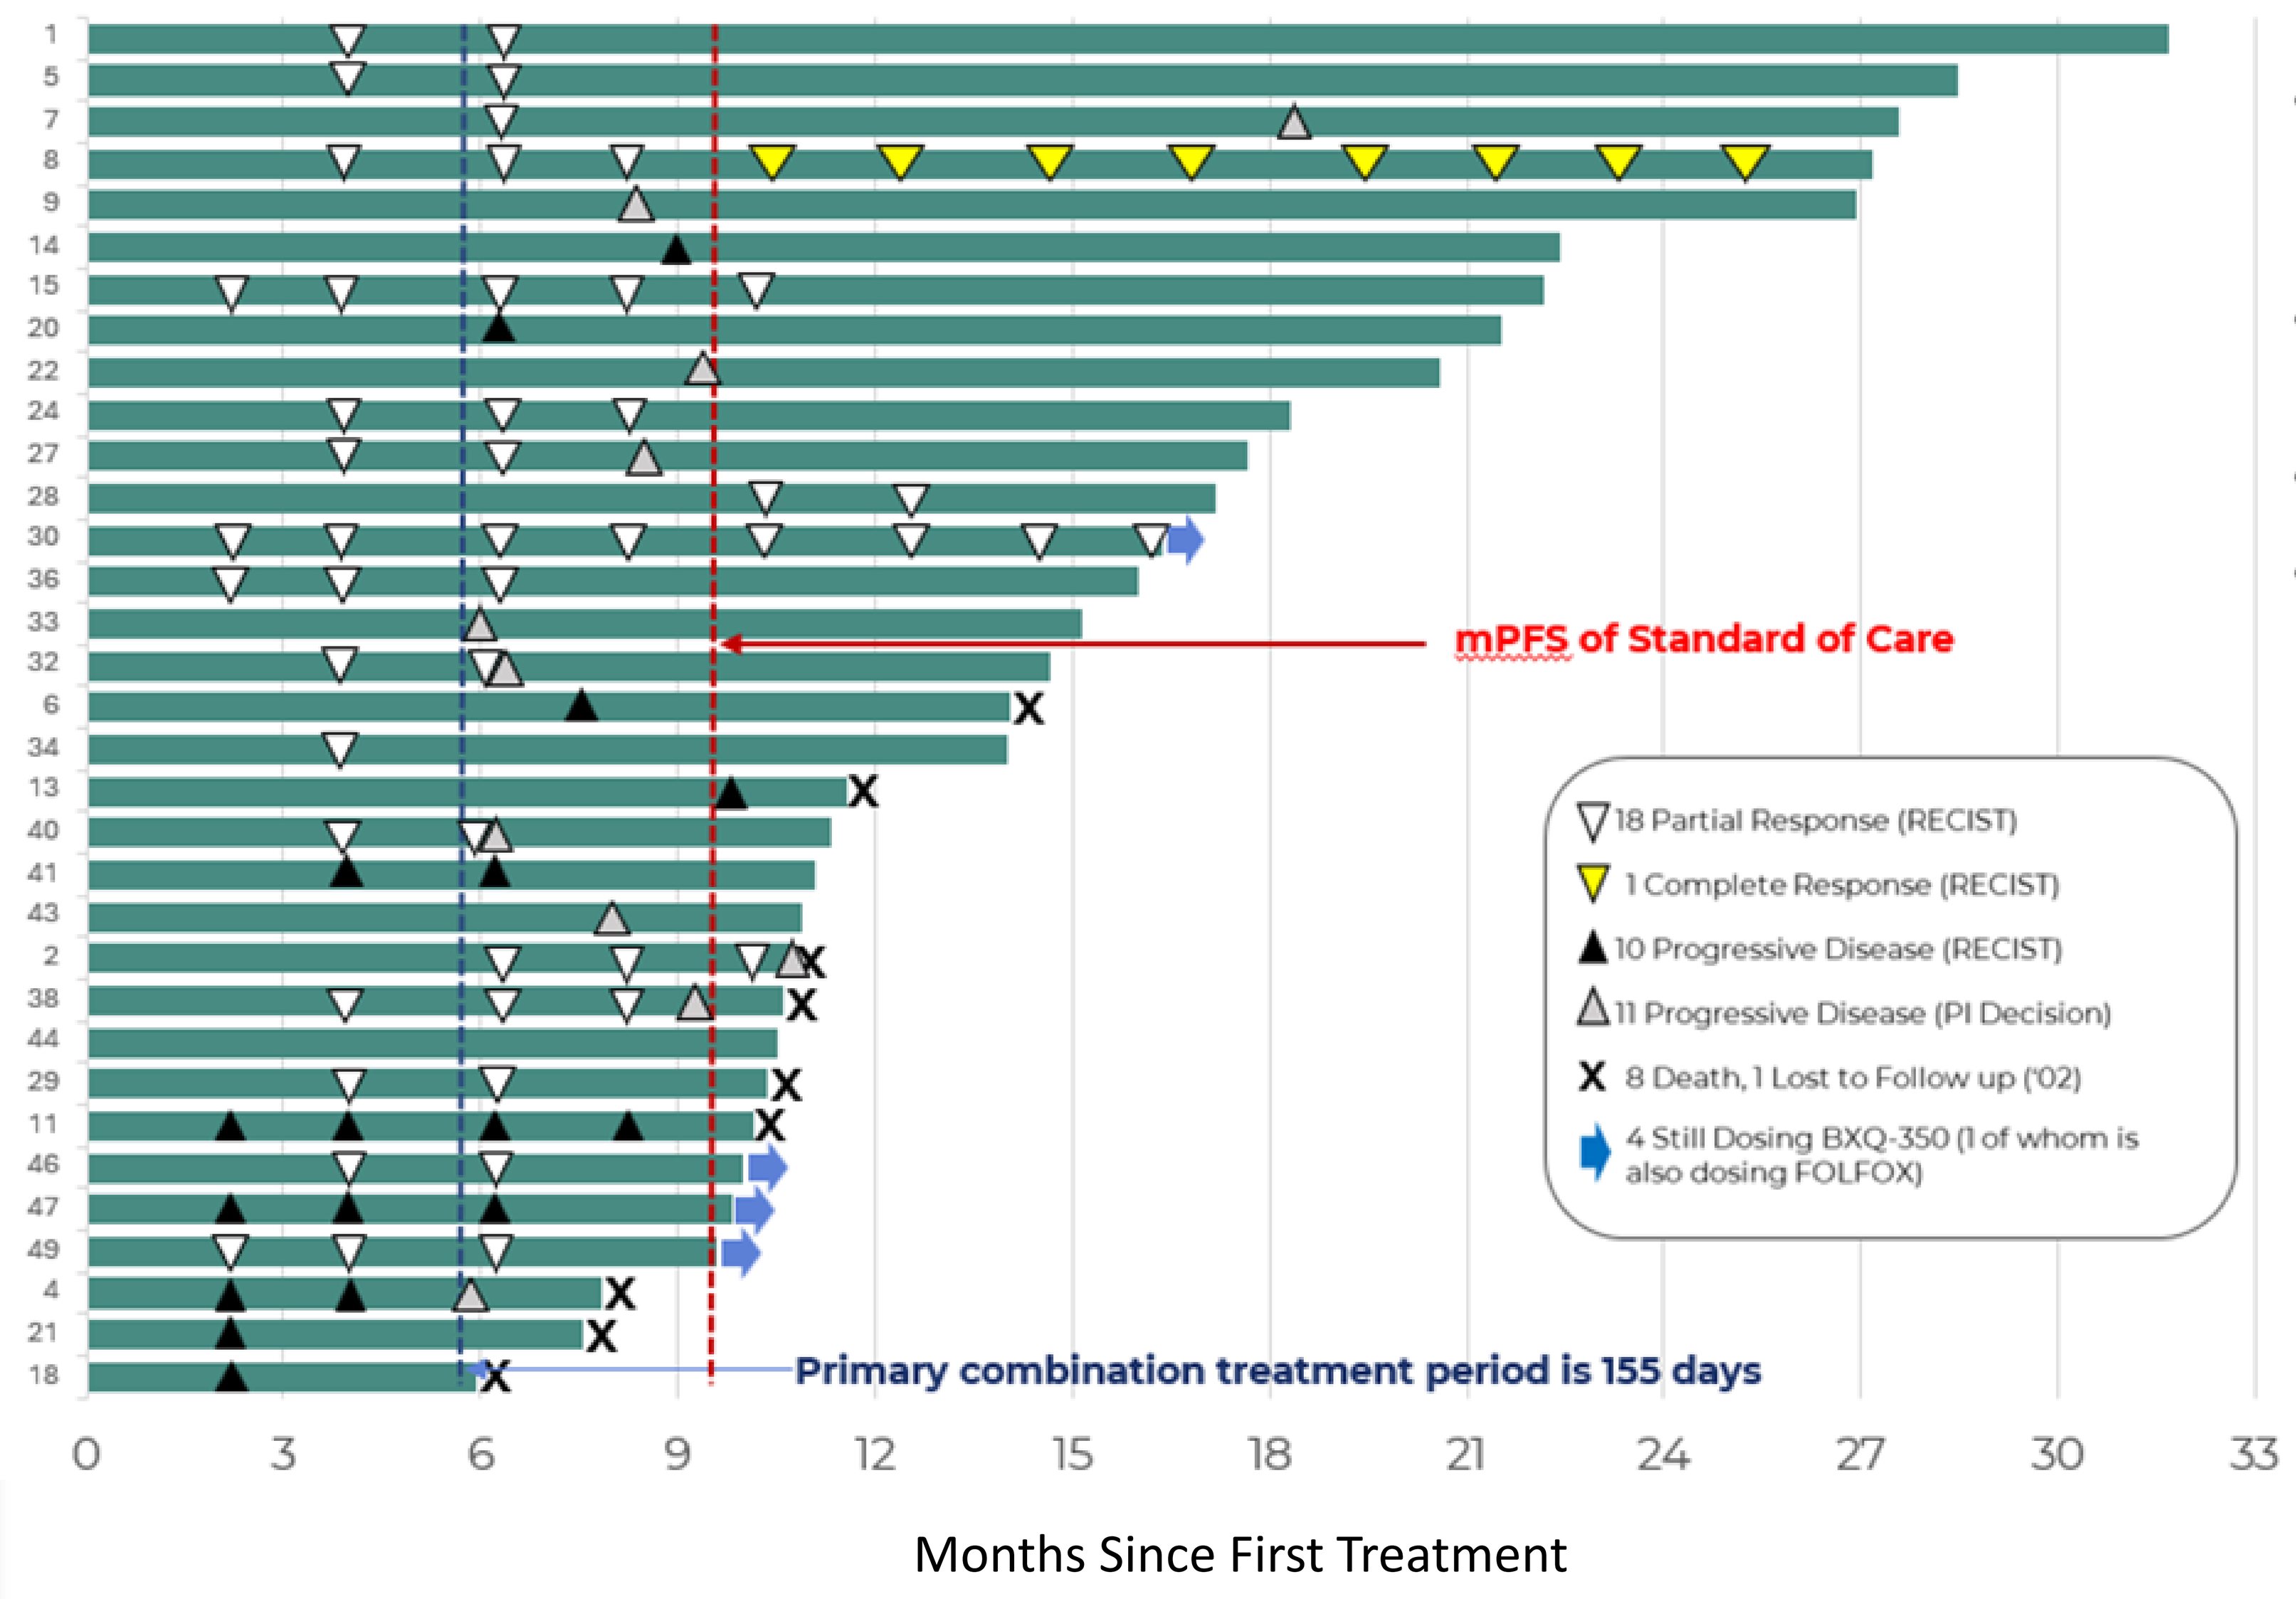

1
5
7
8
9
14
15
20
22
24
27
28
30
36
33
32
6
34
13
40
41
43
2
38
44
29
11
46
47
49
4
21
18
mPFS of Standard of Care
18 Partial Response (RECIST)
1 Complete Response (RECIST)
10 Progressive Disease (RECIST)
11 Progressive Disease (PI Decision)
8 Death, 1 Lost to Follow up ('02)
4 Still Dosing BXQ-350 (1 of whom is also dosing FOLFOX)
Primary combination treatment period is 155 days
0 3 6 9 12 15 18 21 24 27 30 33
Months Since First Treatment

- Primary treatment period of BXQ-350 contemporaneous with mFOLFOX7 – 155 Days
- Pts 1, 2 dosed at 1.8 mg/kg, remainder at RP2D of 2.4 mg/kg
- **ORR D113 19/32 = 59%***
- Subject '008:
 - Oxaliplatin dosing completed D155
 - 2 years dosing BXQ-350
 - Complete response on D299 scan

Central scan data as of Aug 7, 2025

* ORR results slightly differ as there is a pending central scan read outstanding on a single patient.